How Big Is A Microsample?

Claire Grant, Michael Spreadborough, Catherine Wilkinson, Emma Flynn, Sarah Kirk
Alderley Park, Mereside, Alderley Park, Macclesfield, Cheshire SK10 4TG

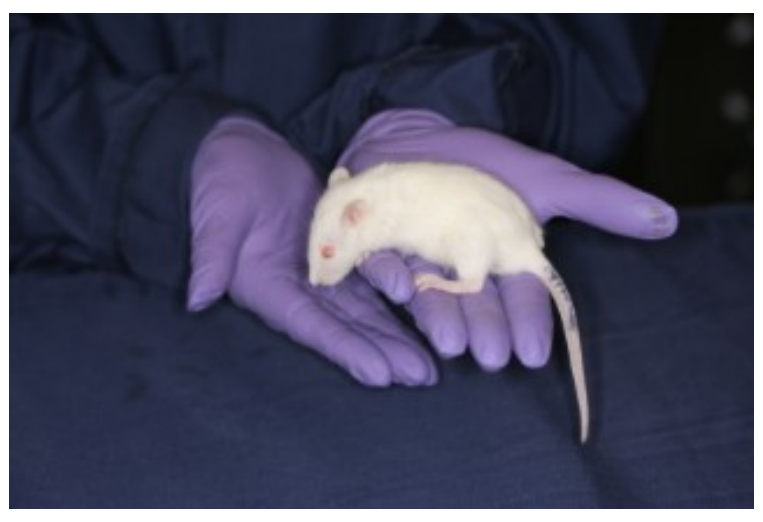

Abstract

Toxicokinetics (TK) is an important endpoint in studies as it allows exposure analysis and calculation of safety margins.
Within AstraZeneca rodent studies a 32 µl microsample is used as standard for TK profiling. A TK profile usually requires at least 6 sampling timepoints within 24 h. The aim of this study was to decrease the microsample volume collected at each timepoint to 20 µl, reducing the overall amount of blood volume taken and the stress involved in the sampling procedure.
The study has shown that a 20 µl microsample does cause less stress to animals during sample collection and exposure analysis is comparable to larger sample volumes. This data will lead to a welfare refinement to study design without impacting on scientific interpretation.

Introduction

Advances in bioanalytical techniques have opened up the potential to use smaller sample volumes (microsamples) to assess drug exposure. A microsample generally refers to a sample of ≤50µl. Microsampling is used in rodent studies, such as those in discovery toxicology.
AstraZeneca uses a 32 µl sampling size as standard. The aim of this study was to show that volume can be reduced to 20 µl.
Animals were dosed with Clonidine, a compound known to reduce heart rate and blood flow. This assessed the sampling procedure in the most difficult circumstances.

Methods

Male Han Wistar rats were sampled collecting 32 or 20 µl microsamples at multiple time points post-dose. Rats (n=8/gp) were dosed orally with vehicle or 0.1 mg/kg Clonidine in a cross over study design. Microsampling was performed at 0.5, 1, 2, 4, 6 and 22 h post-dose.

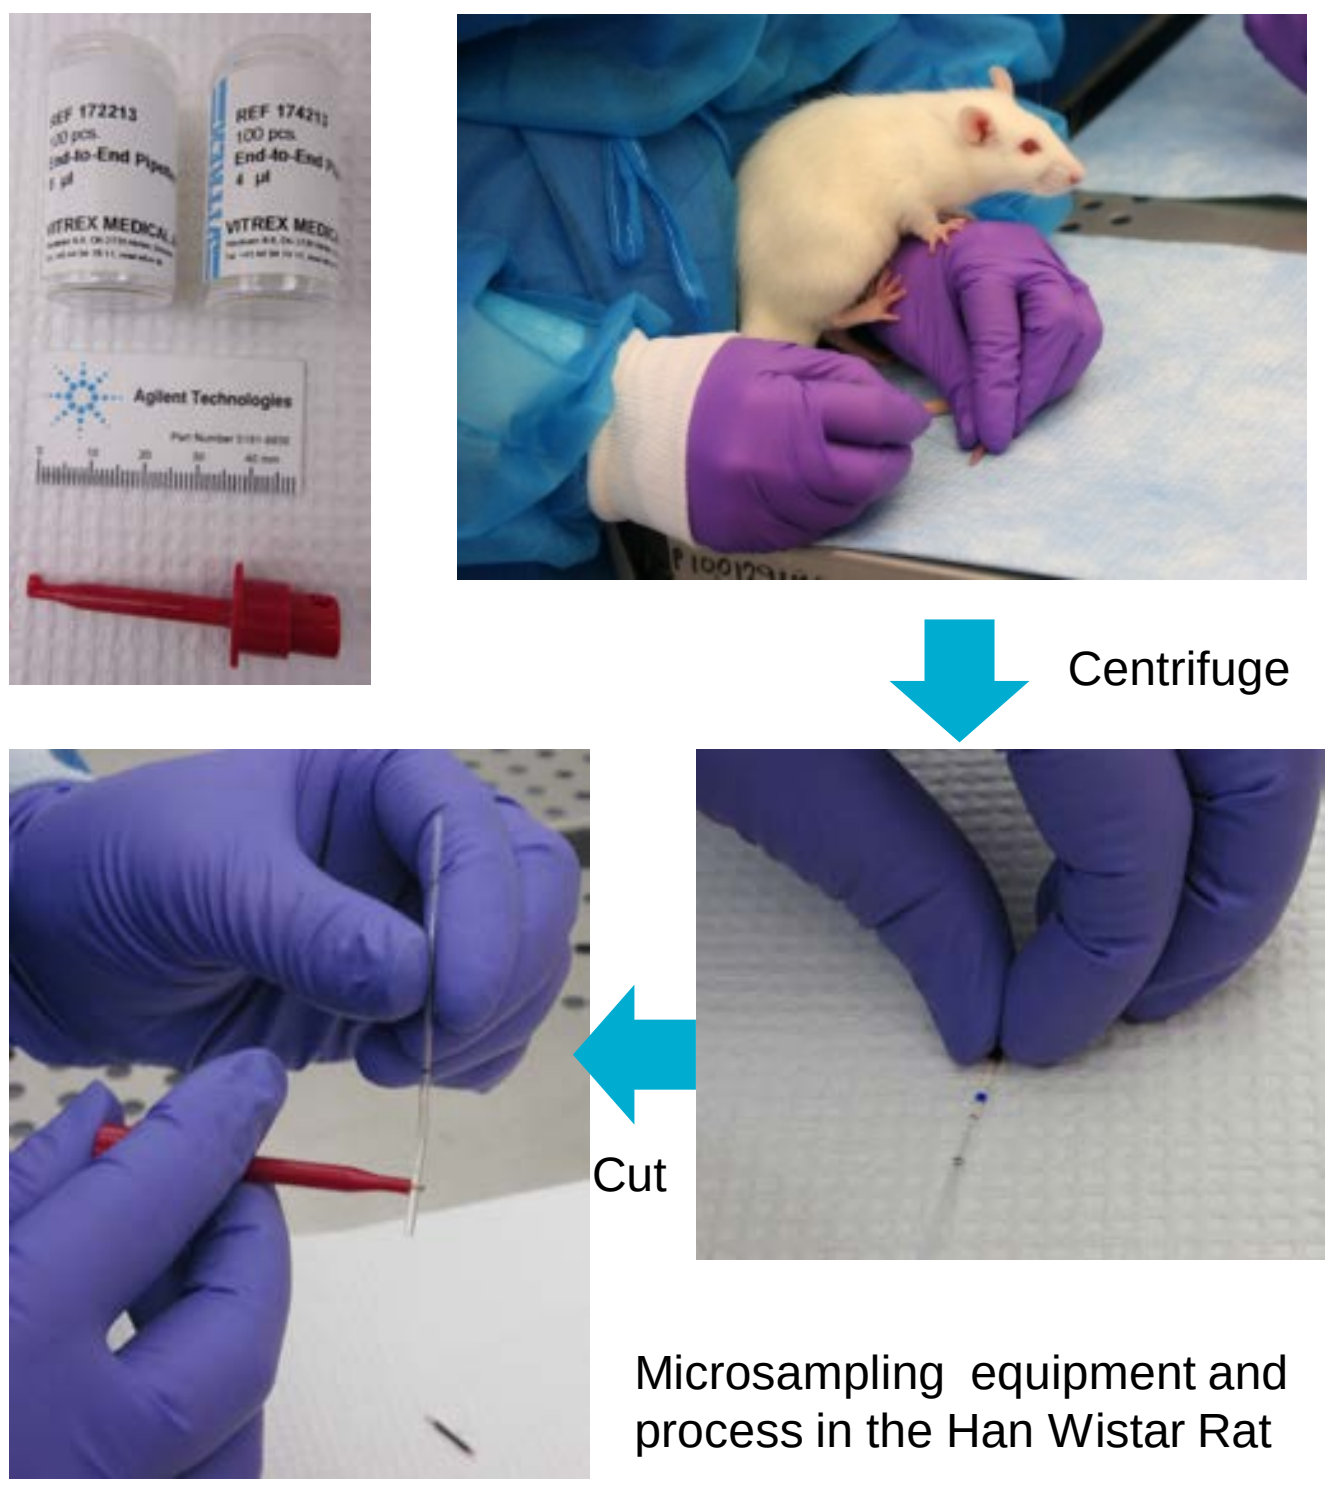
Centrifuge
Cut

Microsampling equipment and process in the Han Wistar Rat

Animals having 32 µL samples were placed in warming chambers for approximately 1 min. 32 µL or 20 µL blood samples were collected into a 75 µL haematocrit capillary tube. Plasma was prepared by centrifugation at 1500g for 10 min at 4°C. The capillary tubes were then cut and plasma harvested using 8 µL capillaries for 32 µl samples or 4 µl capillaries for 20 µl samples and frozen at -20°C. During microsampling animals were minimally restrained by hand and blood was collected from a tail prick site near to, or at the tip of, the tail.

Animals were surgically prepared with telemetry devices (DSI® PhysioTel) and heart rate and blood pressure was recorded for 24 h. Data was analysed (an average of 5 min) before sampling (-20 and -5 min) and after sampling (5, 10, 15, 30, 60 min) as an indicator of the stress experienced by the animals and to assess recovery time.

Plasma samples were analysed for Clonidine concentrations. Following microsample dilution and protein precipitation samples were injected onto a gradient UHPLC system with tandem mass spectrometric detection.

Results

Complete samples were collected at all planned timepoints, except in one animal at 24 h post-dose.
The plasma volume separated from the 20 µl microsamples was assessed and at every sampling timepoint 8 µl of plasma was collected. This indicates that an accurate 4 µl plasma sample could be regularly achievable when collecting a 20 µl microsample in future studies.
Sampling time was quicker, tails required less milking and animals showed less distress (vocalisation and movements) during the 20 µl TK sampling profile compared with the 32 µl sampling.

Clonidine at 0.1 mg/kg caused the expected decrease in heart rate and blood pressure.

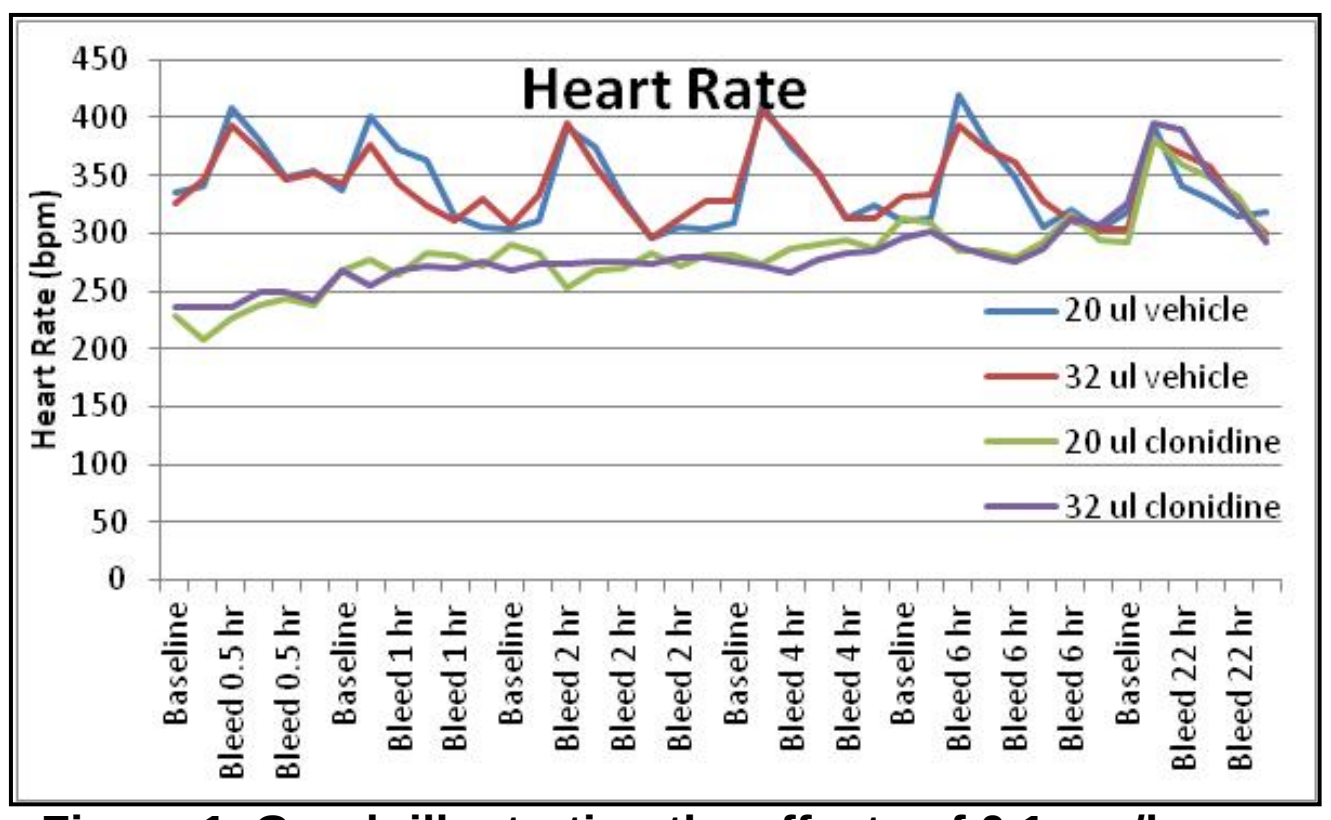
Heart Rate
Heart Rate (bpm)
20 ul vehicle
32 ul vehicle
20 ul clonidine
32 ul clonidine
Baseline
Bleed 0.5 hr
Bleed 1 hr
Bleed 2 hr
Bleed 4 hr
Bleed 6 hr
Bleed 22 hr

Figure 1 Graph illustrating the effects of 0.1 mg/kg Clonidine on heart rate in Han Wistar rats

Blood pressure analysis shows the pressure in animals undergoing 20 µl microsample TK profiles was lower than in the 32 µl microsample TK profiles, indicating the animals were less stressed.

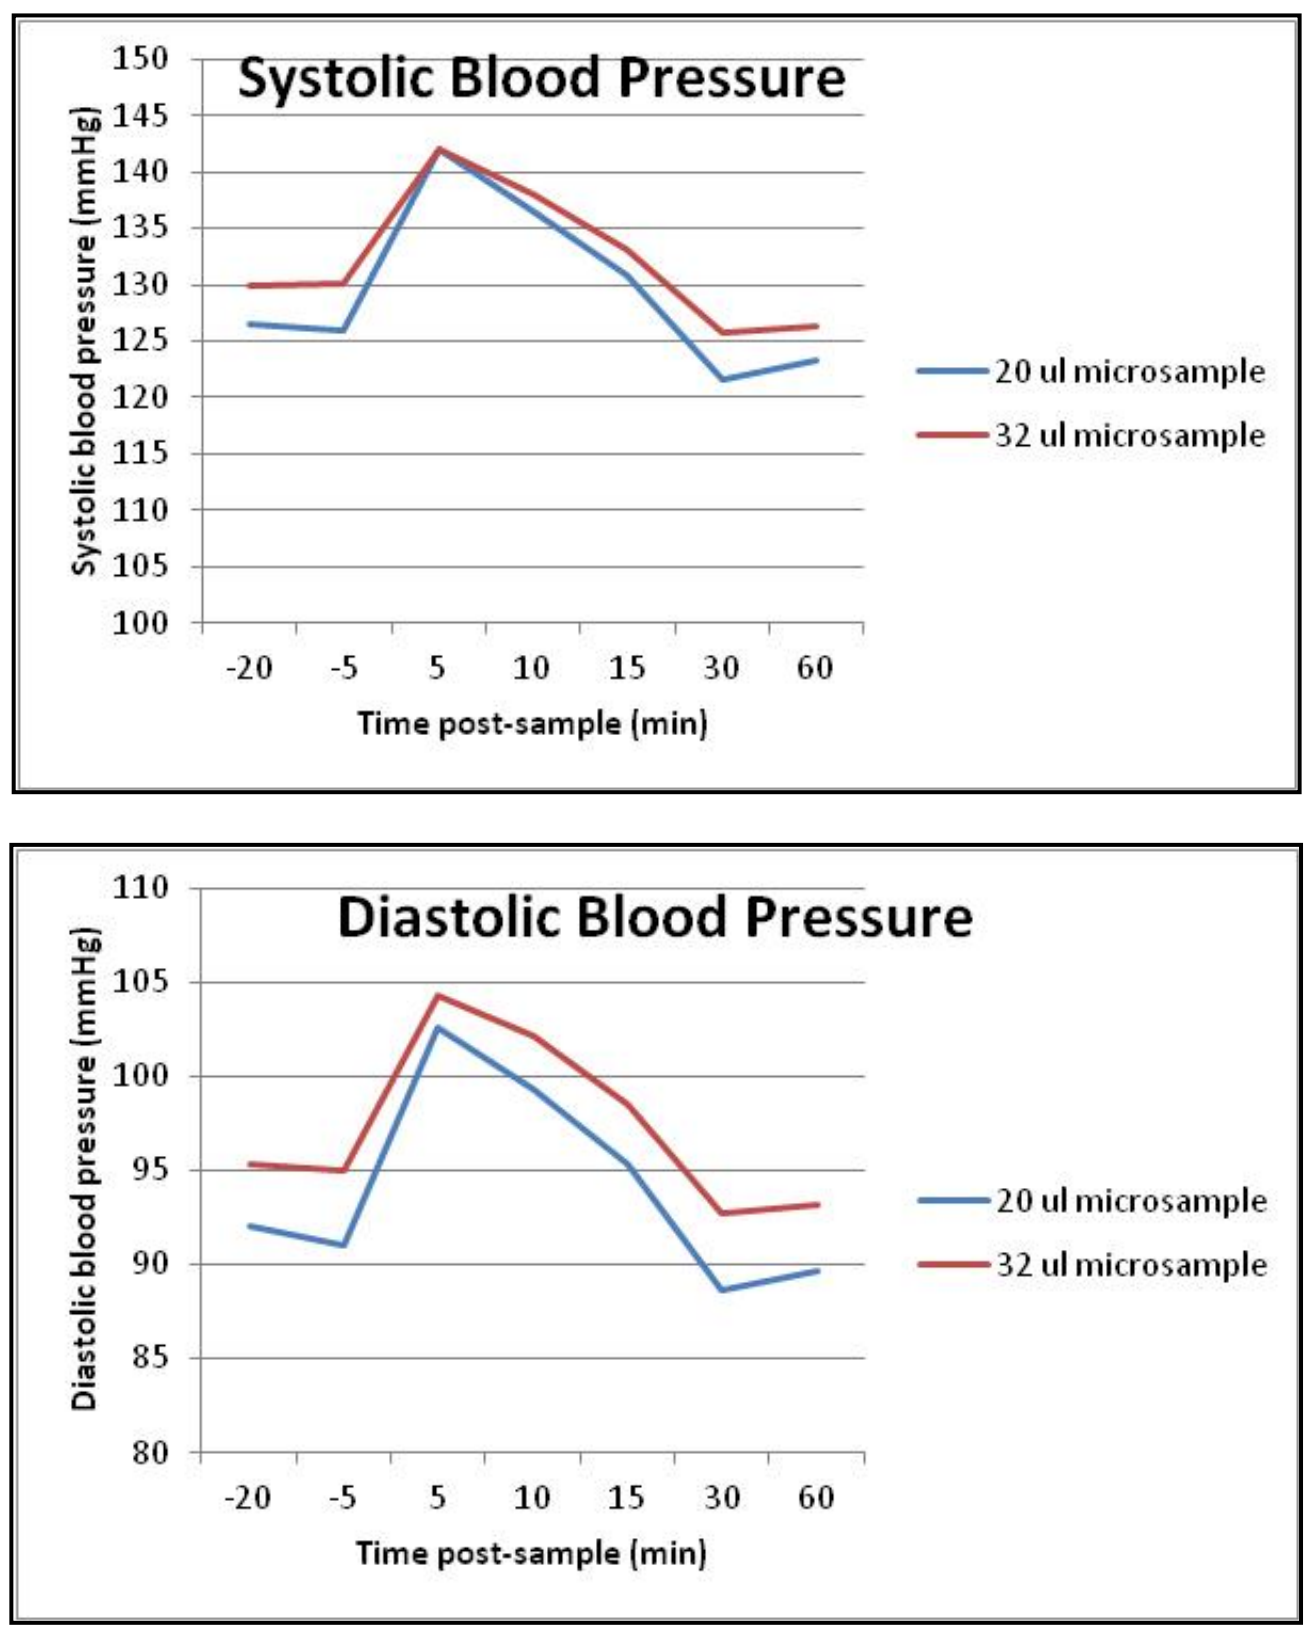
Systolic Blood Pressure
Systolic blood pressure (mmHg)
Time post-sample (min)
20 ul microsample
32 ul microsample
Diastolic Blood Pressure
Diastolic blood pressure (mmHg)
Time post-sample (min)
20 ul microsample
32 ul microsample

Figure 2 Graph showing impact of 20 or 32 µl microsamples on systolic and diastolic blood pressure in vehicle rats.

Mean Clonidine exposure, following oral administration of 0.1 mg/kg Clonidine, was comparable across the sampling time-course. The TK profiles were unaffected by the sample size.
Data was within the normal variability observed for this dataset.

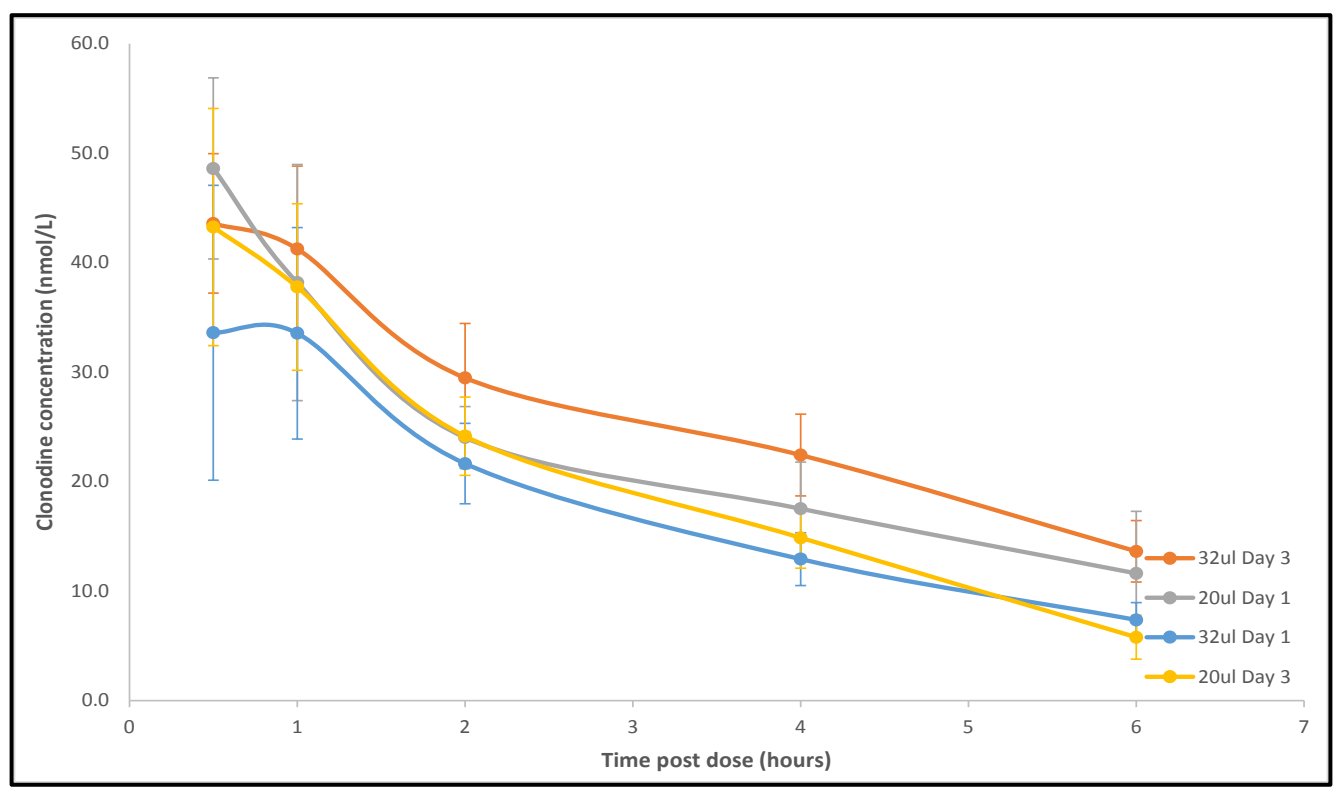
Clonodine concentration (nmol/L)
Time post dose (hours)
32ul Day 3
20ul Day 1
32ul Day 1
20ul Day 3

Figure 3 Graph showing Clonidine exposure data using 20 or 32 ul sampling methods

Conclusions

- AstraZeneca Discovery Toxicology studies will be able to use a TK blood microsample size of 20 µl in the future.
- For a 6 sample TK profile 120 µl of blood will be collected using 20 µl microsamples, this is a reduction of 72 µl per TK profile.
- This is a refinement to current standard practices as the overall blood volume taken is removed and method does not require animal warming.
- The TK exposure data is comparable to larger sampling sizes.
- More data can be obtained from the same animal, as blood could be taken for a more comprehensive TK profile, or can be used for other assessments such as haematology or clinical chemistry.
- This will lead to a large impact for mouse studies. Mice have a much smaller circulating blood volume. Only 300 µl of blood can be collected in a 28 day period in a 25 g mouse, therefore using 20 µl microsamples 2 TK profiles can be collected.
- In future studies a decision will be made depending on study design and in conjugation with the bioanalysis group on the microsampling volume to be used.

Other Activities

Within the last year there have been other investigations into the use of microsampling in studies including:

- Microsampling in rat telemetry studies
- Impact of microsampling on haematology parameters
- Microsampling from subsections of groups

Activities currently being investigated including:

- Microsampling with free roaming intravenous infusions
- Microsampling from main group mice in studies investigating compound effects on reproduction

Acknowledgements
Many thanks to all the people who contributed to the conduct and interpretation of the study.

Supported by AstraZeneca IMED Biotech Unit

Presented at LASA Winter Meeting 2015