Rodent Big Brother: A Comparison to the Modified Irwin Test for Assessing Drug-Induced Changed in Activity and Temperature in Rats

Karen Tse[1], Amy Keerie[1], Rowland Sillito[2], Rachel Collier[1], Claire Grant[3], Catherine Vickers[4], Kathryn Chapman[4], J Douglas Armstrong[2], Will S Redfern[1]

Correspondence: will.redfern@astrazeneca.com

AstraZeneca, Fleming Building, Babraham Institute, Cambridge, CB22 3AT[1], Actual Analytics Ltd, Wilkie Building, Edinburgh, EH8 9AG[2], AstraZeneca, Alderley Park, Cheshire, SK10 4TG[3], NC3Rs, Gibbs Building, 215 Euston Road, London, NW1 2BE[4]

0088

Introduction

- The modified Irwin Test (mIT)[1] is a global neurobehavioural assessment recommended by ICH S7A and involves an in-depth evaluation of each animal at specific timepoints. This however only gives 'snapshots' of behaviours, requires disturbing the animals and is usually only carried out during the light phase.
- Rodent Big Brother (RBB, ActualHCA™, Actual Analytics, UK) continuously monitors ambulatory activity and subcutaneous temperature of individual rats when group-housed in their home cage through a subcutaneously implanted temperature-sensing passive radiofrequency identification (RFID) transponder.
- The objective was to use RBB to evaluate the effects of chlorpromazine and compare it with mIT.

Methods

- Han Wistar rats (200-275g) were implanted with RFID transponders (Biothermo13, Biomark) subcutaneously in the ventral midline region (Fig 1) and housed in groups of 3/cage with a 12h:12h light cycle (lights on at 07:00h)

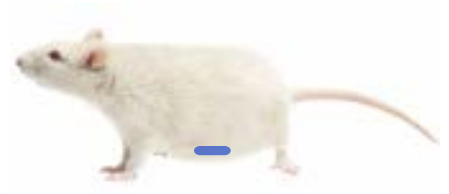

Figure 1. Location of ventral midline RFID transponder

- Chlorpromazine (CPZ; 30 mg/kg po) or vehicle were dosed to animals (n=6/group) in 3 separate environmental conditions:
 - **'Light phase'**; dosed at 09:00h
 - **'Cage change'**; dosed at 09:00h, cage change at T_{max} (2h post-dose)
 - **'Dark phase'**; dosed at 20:00h
- Cages were randomised to treatment groups based on the baseline activity data (number of transitions) collected 2 days before dosing.
- Activity data was derived from baseplate readings via a filtering algorithm that prevents duplicate sequential readings being registered as spurious movement.
- Activity data (transitions between adjacent antenna) was correlated with motion detection detected by the side-view camera to verify real activity. Motion detection was captured by pixel movement from each frame of the video footage.
- A manual home cage observation from the video footage (Fig 4) following dosing at 09:00h was carried out at 15, 30 min, 1, 2, 4 and 24h post-dose.
- An independent mIT was conducted at the same time points as the home cage observations and rectal temperature was measured at 1 and 4h post dose.

Results

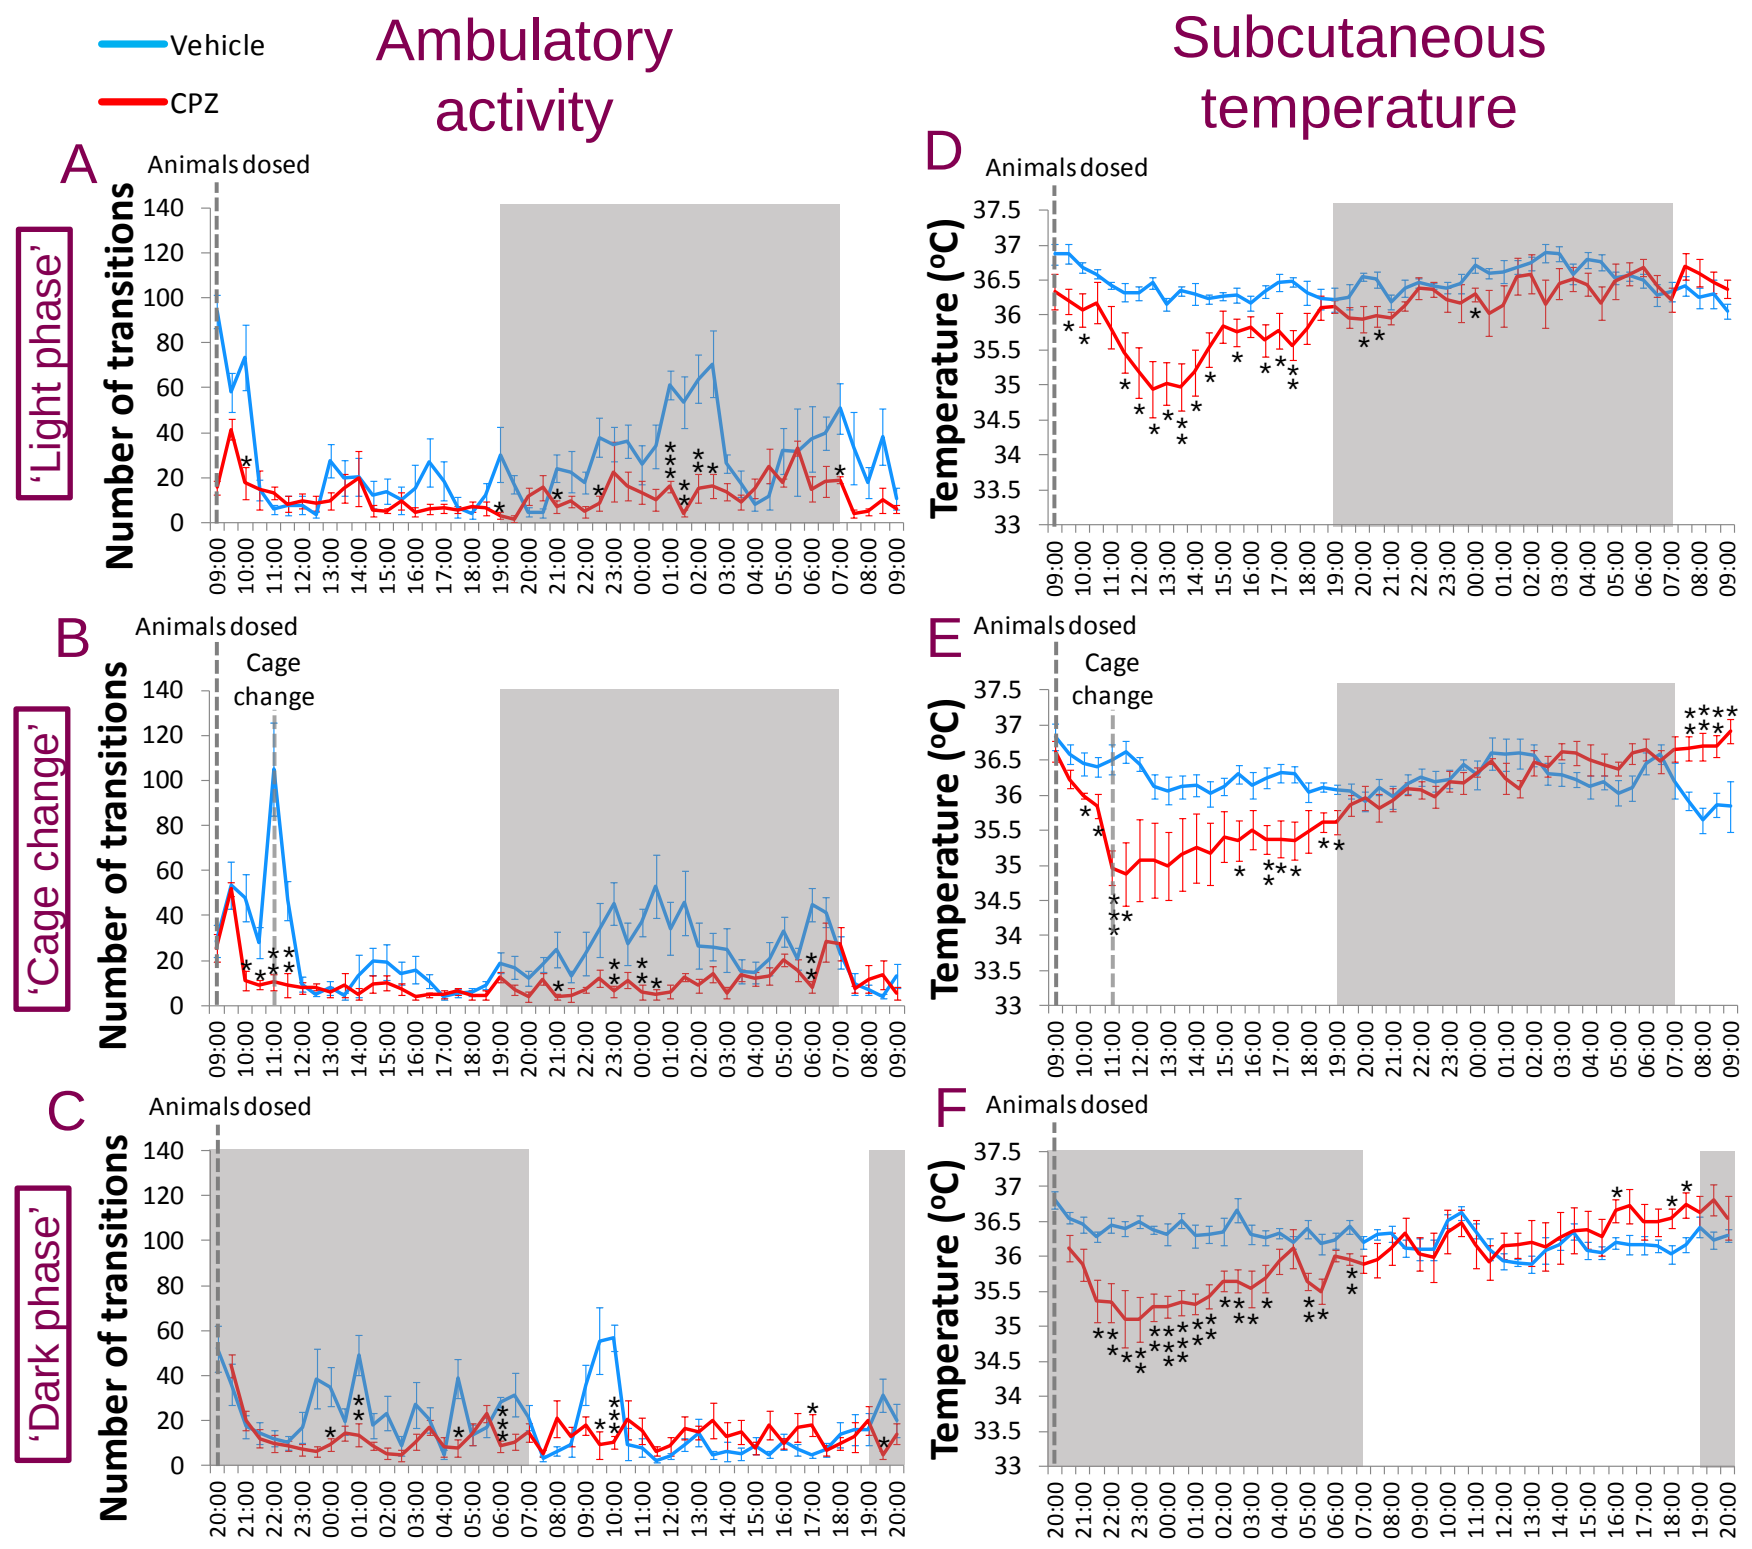
Vehicle
CPZ
Ambulatory activity
Subcutaneous temperature
A
B
C
D
E
F
'Light phase'
'Cage change'
'Dark phase'
Animals dosed
Cage change
Number of transitions
Temperature (°C)

Figure 2. Mean (±SEM) ambulatory activity and temperature from the RFID transponder in CPZ and vehicle treated animals for all 3 separate environmental conditions (n=6/group). Grey shading represents the dark phase (19:00h-07:00h). Dotted lines indicate when animals were dosed and cage changed. Activity and temperature measurements were compared statistically with the vehicle group using AUC and t-test (* *P<0.05; ** P<0.01; *** P<0.001)*

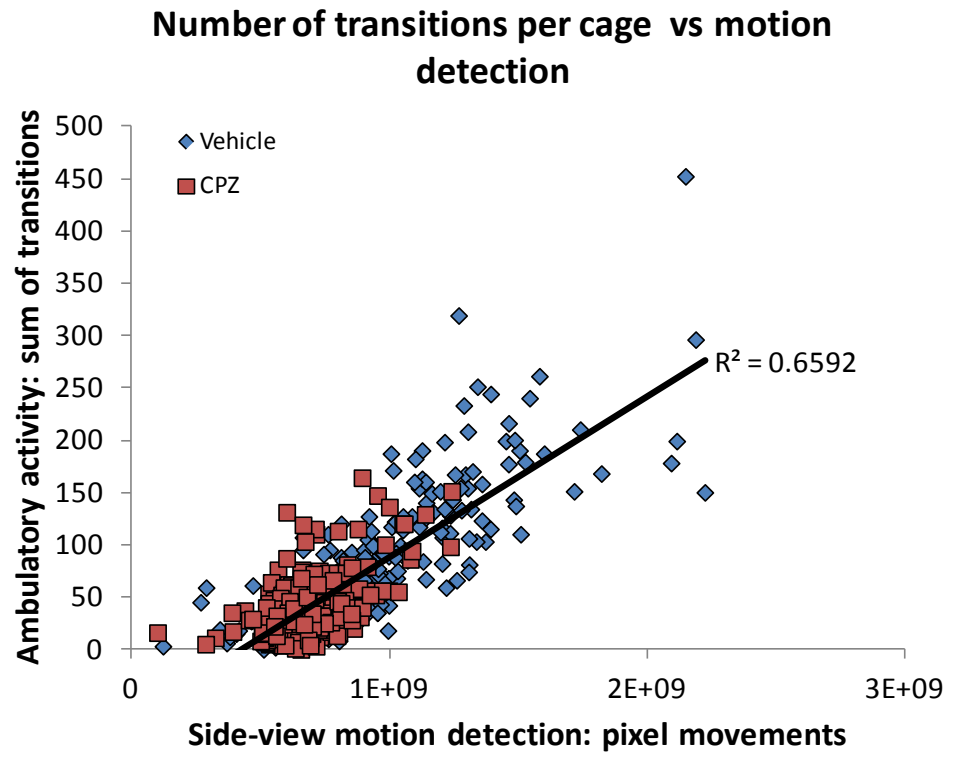
Number of transitions per cage vs motion detection
Vehicle
CPZ
R² = 0.6592
Ambulatory activity: sum of transitions
Side-view motion detection: pixel movements

Figure 3. Motion detection from the side-view HD camera against number of transitions detected by the baseplate for both vehicle and CPZ dosed animals in all 3 phases. Activity detected from the baseplate was correlated with the overall motion detection.

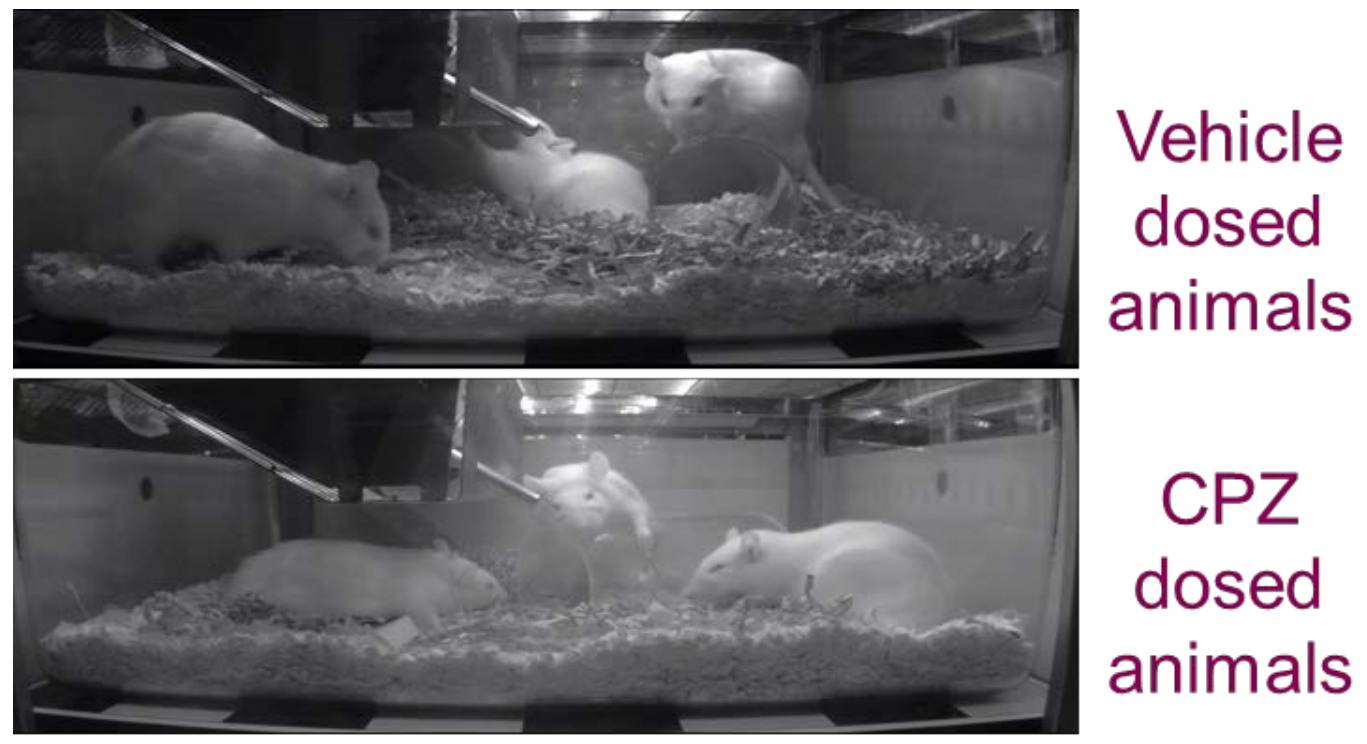
Vehicle dosed animals
CPZ dosed animals

Figure 4. Manual home cage observations from the high quality video footage. Some behaviours can only been observed when the animals move e.g. abnormal gait, or when handled e.g. body tone, and some are not visible from the video e.g. pupil size.

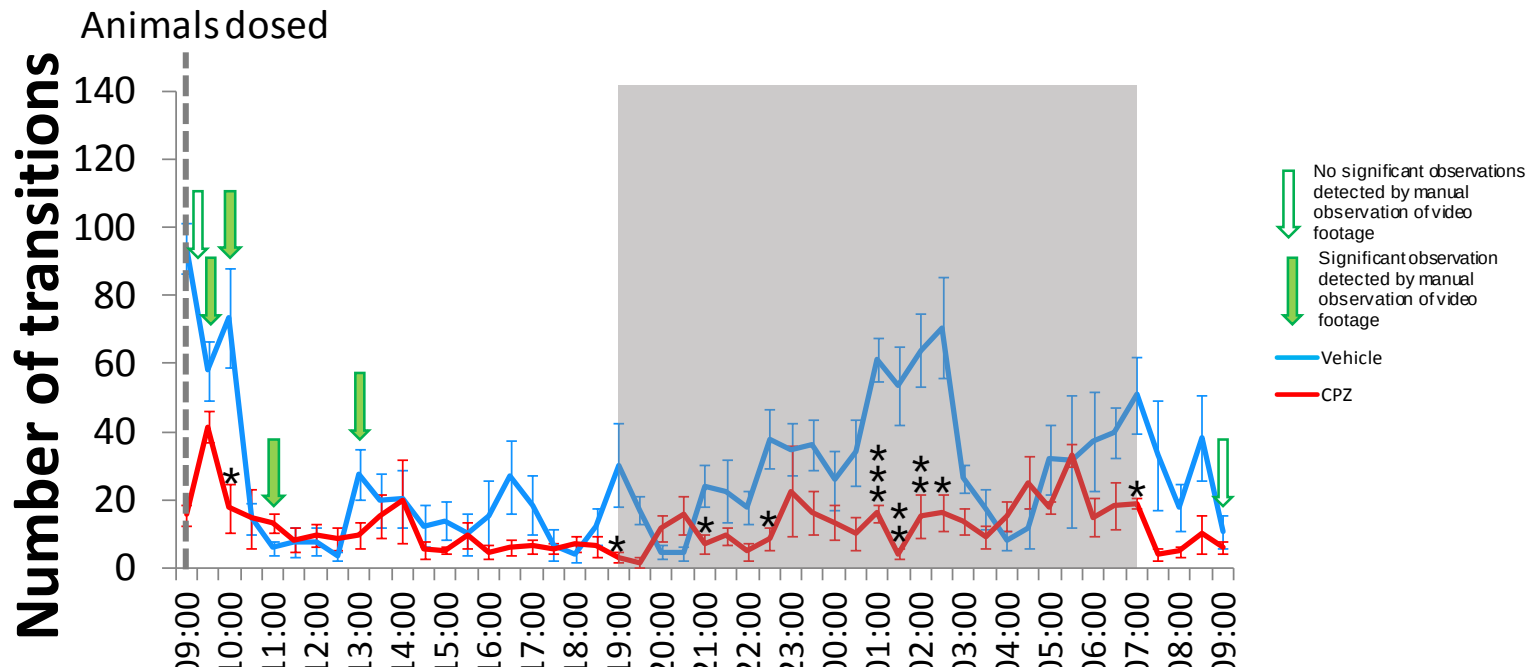
Animals dosed
Number of transitions
No significant observations detected by manual observation of video footage
Significant observation detected by manual observation of video footage
Vehicle
CPZ

Figure 5. Manual home cage observations from the video footage during the light phase compared with the continuous activity data from the RFID transponder. Solid filled arrows indicate significant adverse clinical observation seen, non-filled arrows indicate no significant adverse clinical observations seen. Activity data was compared statistically to vehicle (**P<0.05, **P<0.01, ***P<0.001)*

Chlorpromazine 30mg/kg	Abnormal gait	Abnormal respiration	Piloerection	Spontaneous activity decreased	Temperature
Independent mIT data					Rectal probe
15 min	0/6	0/6	0/6	3/6	
30 min	0/6	0/6	0/6	6/6	
60 min	0/6	0/6	2/6	6/6	-1.3°C
120 min	0/6	0/6	6/6	6/6	
240 min	0/6	0/6	5/6	5/6	-1°C
1440 min	0/6	0/6	0/6	0/6	+0.7°C
Home Cage (video; manual observation)					RFID transponder
15 min	2/6	0/6	0/6	0/6	
30 min	5/6	1/6	0/6	5/6	-0.5°C
60 min	4/6	0/6	0/6	6/6	-0.6°C
120 min	0/6	1/6	0/6	6/6	-0.6°C
240 min	0/6	0/6	0/6	5/6	-0.9°C
1440 min	0/6	0/6	0/6	0/6	+0.3°C

Figure 6. Comparing mIT observations and measurements from an independent experiment, to the manual home cage observations from video footage of animals dosed at the light phase and temperature from the RFID transponder.

Conclusions

- Animals dosed with CPZ (30 mg/kg po) had significantly reduced ambulatory activity (AUC_{0-24h}, P<0.01) and subcutaneous temperature (AUC_{0-12h}, P<0.01) detected by the baseplate compared to the vehicle dosed animals in all three phases.
- CPZ effects on ambulatory activity were more prolonged than was expected from mIT. Effects continued overnight when dosed during the light phase.
- When cage-changed at T_{max}, the pronounced sedative effects of CPZ was revealed during the light phase.
- When dosed during the dark phase, CPZ effects maintained throughout the 24h period.
- CPZ effects on subcutaneous temperature were consistent with the rectal temperature measured in mIT.
- Motion detection data correlated well with the baseplate-derived transition data, verifying the activity measured by the baseplate.
- The HD video captured from the enclosures allows a trained observer to collect home cage observations of individual rats at any time of day or night.
- Decreased spontaneous activity observed was consistent with mIT. Piloerection was not observed from the video footage.
- Future studies will investigate using RBB to detect the effects of a stimulant on rats.

References

[1]Irwin 1968 Psychopharmacologia 13, 222-257

Acknowledgements

I would like to thank the Laboratory Animal Sciences UK group and Mark Piling from the statistics group at AstraZeneca for their support on this study.

Supported by AstraZeneca

Presented at the SPS/JSPS/CSPT Joint Meeting, Vancouver, 18-20 September 2016